Poster #

ARTISTRY-1 study

Phase 2 Study of Switch to Daily BIC + LEN in Individuals on a Complex HIV Treatment Regimen

Karam Mounzer[1], Jihad Slim[2], Moti Ramgopal[3], Malcolm Hedgcock[4], Mark Bloch[5], Jorge Santana[6], Ines Mendes[7], Ying Guo[7], Priyanka Arora[7], Jairo M. Montezuma-Rusca[7], Hal Martin[7], Peter Sklar[7], Conor Moran[8]*, Jared Baeten[9], Sorana Segal-Maurer[10]

[1]Philadelphia FIGHT, Philadelphia, PA, USA; [2]New York Medical College, Valhalla, NY, USA; [3]Midway Immunology and Research Center, Fort Pierce, FL, USA; [4]Spectrum Health, Vancouver, BC, Canada; [5]Holdsworth House Medical Practice, Darlinghurst, NSW, Australia; [6]University of Puerto Rico, San Juan, Puerto Rico; [7]Gilead Sciences, Inc., Foster City, CA, USA; [8]Gilead Sciences Ireland UC, Dublin; [9]Gilead Sciences Ltd, London, UK; [10]New York-Presbyterian Queens, Flushing, NY, USA

*Listed as author for presentation purposes only with permission of all authors

Conclusions

- In this Phase 2 part of the ARTISTRY-1 study:
 - BIC + LEN was highly effective in maintaining viral suppression in participants switching from a complex regimen
 - BIC + LEN was well tolerated, with similar safety profiles observed in the two BIC + LEN treatment groups
- These data support the continued evaluation of a combination of BIC and LEN to optimise treatment in VS PWH who are receiving complex regimens
- A BIC 75 mg/LEN 50 mg STR will be assessed in the Phase 3 part of the study
 - The selected dose of LEN was chosen based on the totality of safety, efficacy, and pharmacokinetic data

Plain Language Summary

- About 8 in every 100 people with HIV take multiple tablets every day to treat their HIV
- A combination of bictegravir plus lenacapavir taken once daily is being tested to help people with HIV reduce the number of tablets that they have to take each day
- In this study, people who were taking multiple tablets every day for their HIV were randomly chosen to either:
 - Receive individual tablets of bictegravir and lenacapavir (at one of two different doses), to be taken together
 - Or to stay on their existing treatment
- After 24 weeks of treatment, people who switched to bictegravir plus lenacapavir still had undetectable levels of HIV in their blood and had few side effects
- This study supports research of a single-tablet combination of bictegravir and lenacapavir in people with HIV

Introduction

- While single tablet regimens (STRs) are the global standard for HIV treatment,[1] approximately 8% of people with HIV (PWH) take complex treatment regimens due to drug resistance, intolerance, toxicity, drug-drug interactions, or contraindications to existing STRs[1-4]
- The combination of bictegravir (BIC) and lenacapavir (LEN) could optimise treatment in virologically suppressed (VS) PWH not eligible for treatment with STRs, or in those for whom STR treatment is suboptimal
 - BIC is an integrase strand transfer inhibitor (INSTI) with a high barrier to resistance[5]
 - LEN is a first-in-class capsid inhibitor, expected to have an absence of resistance in unexposed PWH[6]
- ARTISTRY-1 (NCT05502341) is a randomised, open-label, multicenter, operationally seamless Phase 2/3 study evaluating the efficacy and safety of switching from complex antiretroviral therapy (ART) regimens to BIC and LEN among VS PWH

Objective

- To evaluate the efficacy and safety of switching to a BIC + LEN regimen (BIC 75 mg + LEN 25 mg or BIC 75 mg + LEN 50 mg) versus continuing on stable baseline regimen (SBR) at Week 24 in VS PWH

Methods

- ARTISTRY-1 (NCT05502341) is an ongoing, randomised, open-label, multicenter Phase 2/3 study

Study Design of Phase 2 of ARTISTRY-1

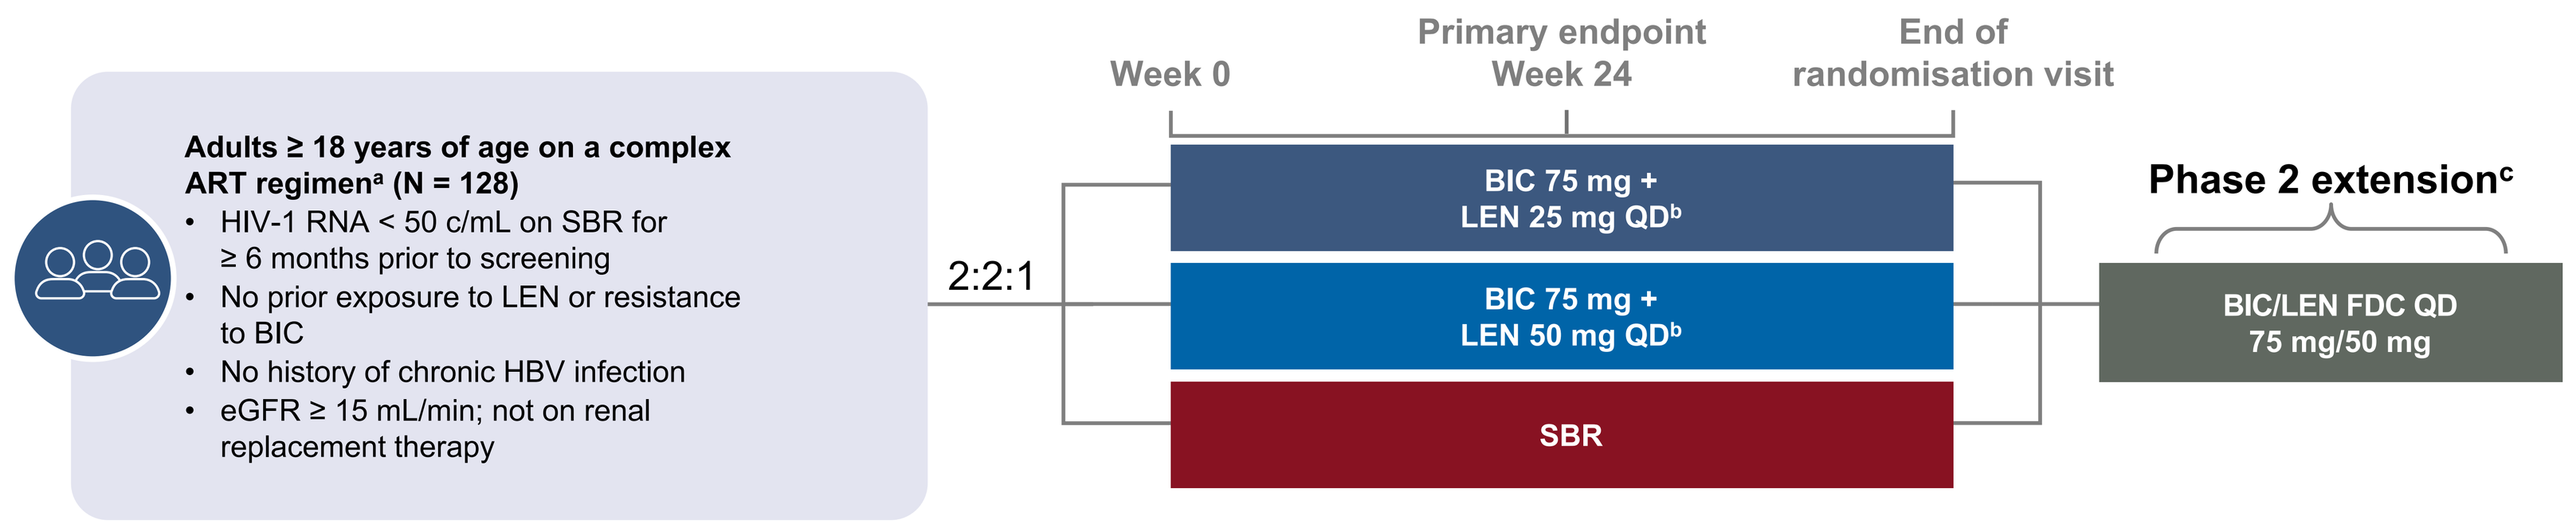

Week 0
Primary endpoint Week 24
End of randomisation visit
Adults ≥ 18 years of age on a complex ART regimen[a] (N = 128)
• HIV-1 RNA < 50 c/mL on SBR for ≥ 6 months prior to screening
• No prior exposure to LEN or resistance to BIC
• No history of chronic HBV infection
• eGFR ≥ 15 mL/min; not on renal replacement therapy
2:2:1
BIC 75 mg + LEN 25 mg QD[b]
BIC 75 mg + LEN 50 mg QD[b]
SBR
Phase 2 extension[c]
BIC/LEN FDC QD 75 mg/50 mg

- Primary endpoint: Proportion of participants with HIV-1 RNA ≥ 50 c/mL at Week 24 determined using the US FDA-defined Snapshot algorithm
- Secondary endpoints:
 - Proportion of participants with HIV-1 RNA < 50 c/mL at Week 24, determined using the US FDA-defined Snapshot algorithm
 - Change from baseline in CD4 cell count at Week 24
 - Proportion of participants with TEAEs up to Week 24
 - Pharmacokinetics of BIC and LEN

[a]Due to viral resistance, intolerance, or contraindication to existing STRs.
[b]All participants receiving BIC + LEN received an oral loading dose of LEN 600 mg on Days 1 and 2 of treatment.
[c]Participants who switch from the SBR regimen in the extension phase will take the oral loading doses of LEN.
ART, antiretroviral therapy; BIC, bictegravir; c, copies; eGFR, estimated glomerular filtration rate; FDA, Food and Drug Administration; FDC, fixed-dose combination; HBV, hepatitis B virus; LEN, lenacapavir; QD, once daily;
SBR, stable baseline regimen; STR, single tablet regimen; TEAE, treatment-emergent adverse event.

- A complex regimen was defined as:
 - A regimen containing a boosted protease inhibitor or a non-nucleoside reverse transcriptase inhibitor plus ≥ 1 other third agent from a class other than nucleos(t)ide reverse transcriptase inhibitors, or
 - A regimen of ≥ 2 pills/day, or a regimen requiring dosing more than once daily, or
 - A regimen containing parenteral agent(s) (excluding a complete long-acting injectable regimen) as well as oral agents
- Efficacy endpoints were analysed using the Full Analysis Set, which included all randomised participants who received ≥ 1 dose of study drug
 - Participants were grouped according to the treatment to which they were randomised
- Safety endpoints were analysed using the Safety Analysis Set, which included all randomised participants who received ≥ 1 dose of study drug
 - Participants were grouped according to the treatment they actually received

Results

Demographic and Baseline Characteristics

	BIC 75 mg + LEN 25 mg N = 51	BIC 75 mg + LEN 50 mg N = 52	SBR N = 25	Total N = 128
Age, years, median (range)	62 (26, 79)	62 (34, 76)	58 (41, 70)	60 (26, 79)
Female at birth, n (%)	13 (25.5)	7 (13.5)	4 (16.0)	24 (18.8)
Race, n (%)				
Asian	2 (3.9)	2 (3.8)	0	4 (3.1)
White	29 (56.9)	34 (65.4)	20 (80.0)	83 (64.8)
Black	18 (35.3)	16 (30.8)	5 (20.0)	39 (30.5)
Other	2 (3.9)	0	0	2 (1.6)
Ethnicity,[a] n (%)				
Hispanic or Latinx	7 (14.0)	9 (17.6)	4 (16.0)	20 (15.9)
Not Hispanic or Latinx	43 (86.0)	42 (82.4)	21 (84.0)	106 (84.1)
HIV-1 RNA ≥ 50 c/mL,[b] n (%)	0	2 (3.8)	0	2 (1.6)
CD4 count, cells/µL, median (Q1, Q3)	583 (460, 764)	624 (517, 791)	585 (285, 733)	610 (435, 766)
CD4 count < 200 cells/µL, n (%)	1 (2.0)	1 (1.9)	2 (8.0)	4 (3.1)
Past medical history of AIDS, n (%)	14 (27.5)	10 (19.2)	2 (8.0)	26 (20.3)
Duration of HIV treatment, years,[c,d] median (Q1, Q3)	27.8 (22.7, 32.4)	27.0 (18.9, 31.5)	26.9 (19.8, 31.9)	27.0 (19.9, 32.0)
Number of prior ARTs, median (Q1, Q3)	4.0 (2.0, 9.0)	7.0 (3.0, 11.0)	8.0 (3.0, 13.0)	6.0 (3.0, 11.0)
Historical resistance mutations,[e] n (%)				
INSTI	0	0	0	0
NNRTI	25 (49.0)	28 (53.8)	14 (56.0)	67 (52.3)
NRTI	31 (60.8)	35 (67.3)	16 (64.0)	82 (64.1)
PI	18 (35.3)	17 (32.7)	11 (44.0)	46 (35.9)
Reasons for taking a complex regimen, n (%)				
History of resistance	44 (86.3)	40 (76.9)	20 (80.0)	104 (81.3)
Intolerance to components of STRs	20 (39.2)	11 (21.2)	7 (28.0)	38 (29.7)
Contraindication to STRs	7 (13.7)	4 (7.7)	1 (4.0)	12 (9.4)
Comorbidities, n (%)				
Dyslipidemia	39 (76.5)	37 (71.2)	20 (80.0)	96 (75.0)
Diabetes mellitus	36 (70.6)	36 (69.2)	16 (64.0)	88 (68.8)
Hypertension	34 (66.7)	29 (55.8)	15 (60.0)	78 (60.9)
Chronic kidney disease	12 (23.5)	13 (25.0)	4 (16.0)	29 (22.7)

[a]Local regulators did not allow collection of ethnicity information for one participant in the BIC 75 mg + LEN 25 mg group and one participant in the BIC 75 mg + LEN 50 mg group. [b]Two participants had screening HIV-1 RNA < 50 c/mL confirmed but had Day 1/baseline HIV-1 RNA ≥ 50 c/mL; these participants had been on SBR for > 10 months and > 46 months, respectively. [c]Duration of HIV treatment = (first dose date – start date of the first HIV treatment + 1 day)/365.25. [d]N = 50 for BIC 75 mg + LEN 25 mg group. [e]Alternative response was no or not available. ART, antiretroviral therapy; BIC, bictegravir; BMI, body mass index; c, copies; INSTI, integrase strand transfer inhibitor; LEN, lenacapavir; NNRTI, non-nucleoside reverse transcriptase inhibitor; NRTI, nucleos(t)ide reverse transcriptase inhibitor; PI, protease inhibitor; Q, quartile; SBR, stable baseline regimen; STR, single tablet regimen.

Diversity of Complex ART Regimens in Phase 2

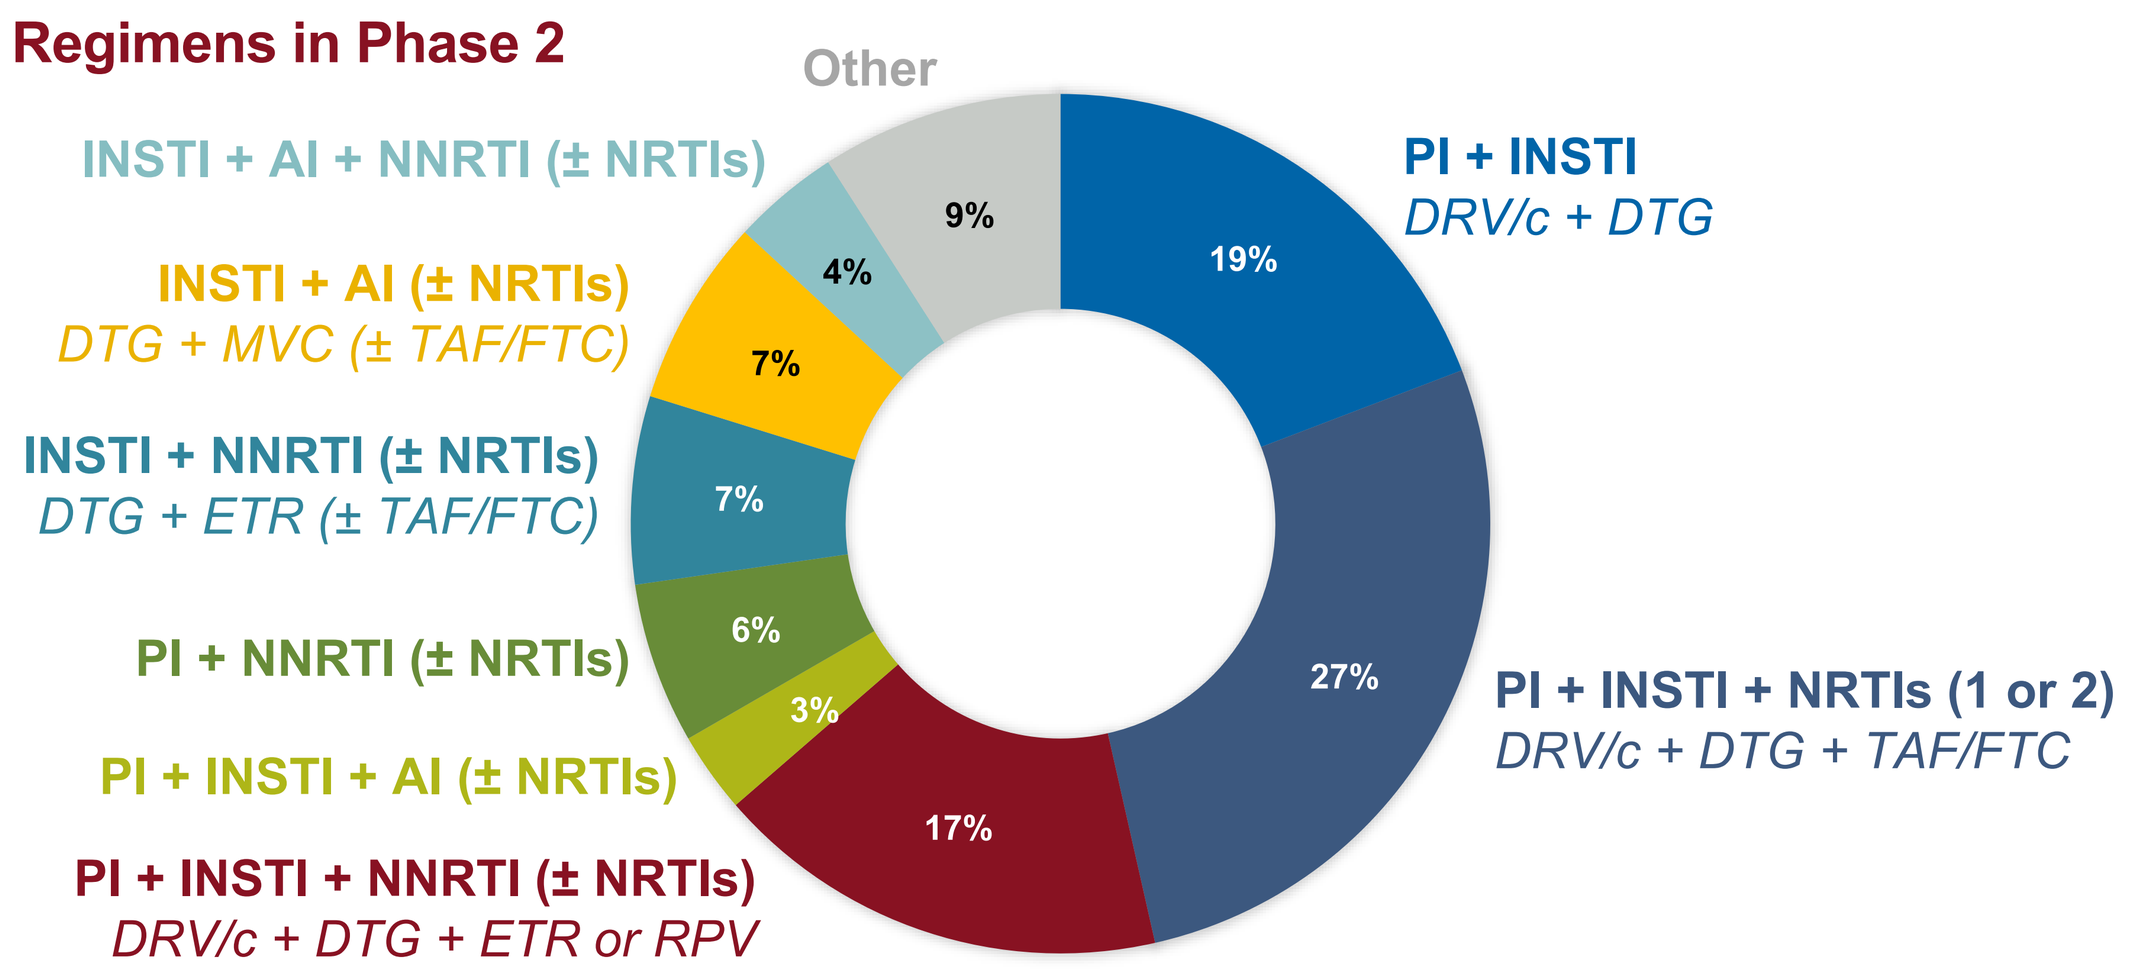

Other
9%
PI + INSTI
DRV/c + DTG
19%
PI + INSTI + NRTIs (1 or 2)
DRV/c + DTG + TAF/FTC
27%
PI + INSTI + NNRTI (± NRTIs)
DRV/c + DTG + ETR or RPV
17%
PI + INSTI + AI (± NRTIs)
3%
PI + NNRTI (± NRTIs)
6%
INSTI + NNRTI (± NRTIs)
DTG + ETR (± TAF/FTC)
7%
INSTI + AI (± NRTIs)
DTG + MVC (± TAF/FTC)
7%
INSTI + AI + NNRTI (± NRTIs)
4%

N = 128. The most common regimen(s) are shown in italics in each regimen category box; this is not an exhaustive list. Percentages do not sum to 100% due to rounding. AI, attachment inhibitor; DRV/c, darunavir/cobicistat; DTG, dolutegravir; ETR, etravirine; FTC, emtricitabine; INSTI, integrase strand transfer inhibitor; MVC, maraviroc; NNRTI, non-nucleoside reverse transcriptase inhibitor; NRTI, nucleos(t)ide reverse transcriptase inhibitor; PI, protease inhibitor; RPV, rilpivirine; TAF, tenofovir alafenamide.

Results

Complexity of ART Regimens at Baseline[a] in Phase 2

	BIC 75 mg + LEN 25 mg N = 51	BIC 75 mg + LEN 50 mg N = 52	SBR N = 25	Total N = 128
Number of pills/day, median (range)	2.0 (2.0, 8.0)	3.0 (2.0, 9.0)	3.0 (2.0, 8.0)	3.0 (2.0, 9.0)
Number of ARTs, median (range)	2.0 (1.0, 5.0)	2.5 (2.0, 5.0)	3.0 (2.0, 5.0)	2.0 (1.0, 5.0)
Dosing frequency of ARTs, n (%)				
Daily	47 (92.9)	46 (88.5)	22 (88.0)	115 (89.8)
Twice per day	18 (35.3)	22 (42.3)	13 (52.0)	53 (41.4)
Other	1 (2.0)	0	0	1 (0.8)

[a]ART use at baseline was defined as the ARTs taken on or up to 14 days prior to Day 1. Multiple reported ARTs were counted only once per participant for each drug name and each drug class.
ART, antiretroviral therapy; BIC, bictegravir; LEN, lenacapavir; SBR, stable baseline regimen.

Virologic Outcome at Week 24 (FDA Snapshot Algorithm)

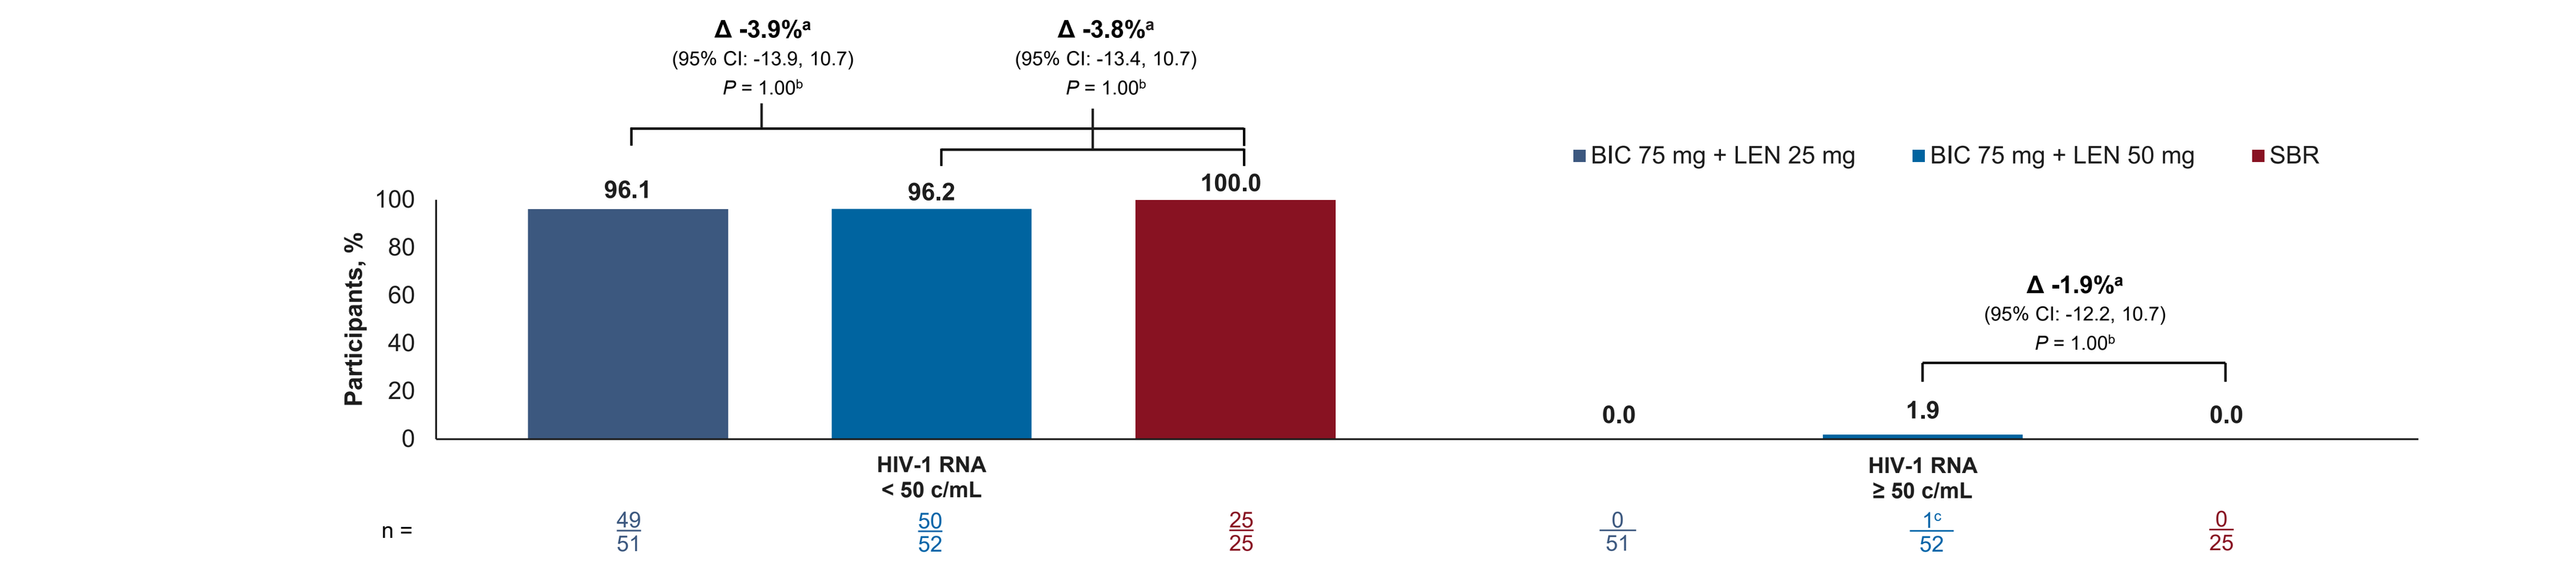

Δ -3.9%[a]
(95% CI: -13.9, 10.7)
P = 1.00[b]
Δ -3.8%[a]
(95% CI: -13.4, 10.7)
P = 1.00[b]
BIC 75 mg + LEN 25 mg
BIC 75 mg + LEN 50 mg
SBR
Participants, %
100
80
60
40
20
0
96.1
96.2
100.0
HIV-1 RNA < 50 c/mL
0.0
1.9
0.0
Δ -1.9%[a]
(95% CI: -12.2, 10.7)
P = 1.00[b]
HIV-1 RNA ≥ 50 c/mL
n =
49/51
50/52
25/25
0/51
1[c]/52
0/25

Two participants (3.9%) in the BIC 75 mg + LEN 25 mg group and one (1.9%) in the BIC 75 mg + LEN 50 mg group had no virologic data in the Week 24 window; reasons: one participant (2.0%) in the BIC 75 mg + LEN 25 mg group and one (1.9%) in the BIC 75 mg + LEN 50 mg group discontinued study drug due to an AE/death and last available HIV-1 RNA < 50 c/mL, and one participant (2.0%) in the BIC 75 mg + LEN 25 mg group discontinued study drug due to other reasons and last available HIV-1 RNA < 50 c/mL. [a]Difference in % (95% CI): BIC + LEN – SBR calculated based on an unconditional exact method using two inverted one-sided tests. [b]Based on Fisher exact test. [c]HIV-1 RNA ≥ 50 c/mL in Week 24 window (later suppressed to < 50 c/mL without regimen change). No genotype/phenotype was performed as virologic failure did not reach threshold as per protocol (> 200 c/mL). AE, adverse event; BIC, bictegravir; c, copies; FDA, Food and Drug Administration; LEN, lenacapavir; SBR, stable baseline regimen.

Change From Baseline in CD4 Cell Count and % at Week 24

	BIC 75 mg + LEN 25 mg N = 51	BIC 75 mg + LEN 50 mg N = 52	SBR N = 25
Change in CD4 cell count, cells/µL			
n	49	51	25
Mean (SD)	33 (155.0)	5 (145.3)	43 (179.6)
Median (Q1, Q3)	18 (-39, 70)	-16 (-80, 93)	42 (-36, 90)
Difference in LSM (95% CI): BIC + LEN vs SBR	0 (-71, 70)	-13 (-84, 58)	–
P value: BIC + LEN vs SBR	0.9893	0.7173	–
Change in CD4 percentage, %			
n	49	51	25
Mean (SD)	-0.5 (3.09)	-0.2 (3.07)	0.7 (5.60)
Median (Q1, Q3)	-0.1 (-2.2, 1.3)	-0.6 (-2.4, 2.0)	0.2 (-1.2, 1.6)

Difference in LSM and *P* value were from ANCOVA model of change from baseline CD4 cell count with treatment as fixed effect and baseline CD4 cell count as a covariate. ANCOVA, analysis of covariance; BIC, bictegravir; LEN, lenacapavir; LSM, least squares mean; Q, quartile; SBR, stable baseline regimen.

TEAEs Up to Week 24 – Overall Summary

	BIC 75 mg + LEN 25 mg N = 51	BIC 75 mg + LEN 50 mg N = 52	SBR N = 25
Any TEAE	39 (76.5)	33 (63.5)	17 (68.0)
TEAE Grade 3 or higher	4 (7.8)	2 (3.8)	1 (4.0)
TEAE related to study drug or SBR	9 (17.6)	3 (5.8)	0
TE serious AE	2 (3.9)	1 (1.9)	2 (8.0)
TEAE leading to discontinuation of study drug/SBR	1 (2.0)	1 (1.9)	0
Nausea[a]	1 (2.0)	0	0
Vomiting[b]	0	1 (1.9)	0
TEAE leading to discontinuation of study	1 (2.0)	1 (1.9)	0
Death[c]	0	1 (1.9)	0

Data shown as n (%). N-values represent numbers of participants. Only TEAEs with onset date on or before the nominal Week 24 visit date were included in this summary. One additional unrelated serious AE was reported after Week 24 up to the Week 24 data snapshot date on July 12, 2023, for each BIC/LEN group (anxiety n = 1; coronary artery disease n = 1). There were no serious AEs leading to discontinuation of study up to Week 24. [a]Grade 1 nausea on Day 1. [b]Grade 3 worsening of vomiting in a participant with preexisting episodes of nausea and vomiting. [c]Death unrelated to study drug; cause of death: coronary artery disease. AE, adverse event; BIC, bictegravir; LEN, lenacapavir; SBR, stable baseline regimen; TE, treatment-emergent; TEAE, treatment-emergent adverse event.

TEAEs by Preferred Term Up to Week 24 (Frequency ≥ 5%)

	BIC 75 mg + LEN 25 mg N = 51	BIC 75 mg + LEN 50 mg N = 52	SBR N = 25
Any TEAE	39 (76.5)	33 (63.5)	17 (68.0)
COVID-19	4 (7.8)	2 (3.8)	2 (8.0)
Diarrhoea	5 (9.8)	2 (3.8)	1 (4.0)
Hypertension[a]	0	4 (7.7)	1 (4.0)
Constipation	3 (5.9)	2 (3.8)	0
Cough	3 (5.9)	1 (1.9)	0
Nasopharyngitis	4 (7.8)	0	0

Data shown as n (%). N-values represent numbers of participants. Only TEAEs with onset date on or before the nominal Week 24 visit date were included. All TEAEs with frequency ≥ 5% were Grade 1 or 2 apart from one occurrence of Grade 3 diarrhea (unrelated to study drug). [a]All hypertension events were assessed as unrelated to study drug or SBR; 2 of the 4 participants with hypertension in the BIC 75 mg + LEN 50 mg group had a past medical history of hypertension. BIC, bictegravir; LEN, lenacapavir; SBR, stable baseline regimen; TEAE, treatment-emergent adverse event.

Treatment-Emergent Laboratory Abnormalities Up to Week 24

Maximum Postbaseline Toxicity Grade	BIC 75 mg + LEN 25 mg N = 51	BIC 75 mg + LEN 50 mg N = 52	SBR N = 25
Any Grade 1 or higher	42 (82.4)	39 (75.0)	21 (84.0)
Grade 3	5 (9.8)	9 (17.3)	8 (32.0)
Creatinine clearance low	0	4 (7.7)	3 (12.0)
Creatinine high	0	1 (1.9)	0
Creatine kinase high	1 (2.0)	0	0
Urine glucose (glycosuria)	2 (3.9)	5 (9.6)	1 (4.0)
Fasting serum glucose high	0[a]	2 (3.8)	0
Non-fasting serum glucose high	2 (9.1)[b]	1 (4.3)[c]	1 (6.7)[d]
Lipase high	1 (2.0)	0	0
Total bilirubin high	0	0	1 (4.0)
Fasting total cholesterol high	0[e]	0	2 (8.0)
Fasting triglycerides high	0[e]	1 (1.9)	0
Fasting LDL high	0[e]	0	2 (8.0)
Grade 4	1 (2.0)	3 (5.8)	0
Creatinine clearance low	1 (2.0)	2 (3.8)	0
Lipase high	0	1 (1.9)	0

Severity grades were defined by the Division of AIDS Toxicity Grading Scale, Version 2.1.[7]
Data shown as n (%). N-values represent numbers of participants. Some participants experienced more than one treatment-emergent laboratory anomaly. Grade 3 and 4 creatinine clearance low and urine glucose were reported in participants with chronic kidney disease or diabetes mellitus, respectively. The remaining Grade 3/4 laboratory abnormalities were either consistent with participants' medical history or transient and not clinically significant. [a]N = 50. [b]N = 22. [c]N = 23. [d]N = 15. [e]N = 49. BIC, bictegravir; LEN, lenacapavir; LDL, low-density lipoprotein; SBR, stable baseline regimen.

References: **1.** DHHS. https://clinicalinfo.hiv.gov/sites/default/files/guidelines/documents/adult-adolescent-arv/guidelines-adult-adolescent-arv.pdf (accessed February 1, 2024). **2.** Chang HM, et al. *BMC Infect Dis*. 2022;22:2. **3.** Rolle C-P, et al. *J Virus Erad*. 2020;6:100021. **4**. Gilead Sciences, Inc. Data on file. **5.** Acosta RK, et al. *Antimicrob Agents Chemother*. 2019;63:e02533-18. **6.** Dvory-Sobol H, et al. *Curr Opin HIV AIDS*. 2022;17:15-21. **7.** DHHS. https://rsc.niaid.nih.gov/sites/default/files/daidsgradingcorrectedv21.pdf (accessed February 1, 2024).

Disclosures: **KM** reports payments for participation in advisory boards and speakers' bureaus from Epividian, Gilead Sciences, Inc., Janssen Therapeutics, Merck, and ViiV Healthcare. **JiS:** reports speaker honoraria from AbbVie, Gilead Sciences, Inc., Merck, and ViiV Healthcare. **MR** has no conflicts of interests to report. **MH** reports consulting fees and speaker honoraria from Gilead Sciences, Inc., Merck, and ViiV Healthcare. **MB** has no conflicts of interest to report. **JoS** reports grants from NIH; and consulting fees, payment for lectures, and payment for participation in advisory boards from AbbVie. **IM, YG, PA, JMM, PS,** and **JB** are employees of and own stock in Gilead Sciences, Inc. **HM** was an employee of Gilead Sciences, Inc. and remains a stockholder. **SS-M** reports research grants from Gilead Sciences, Inc.; advisory/consulting fees from Gilead Sciences, Inc., Janssen, Theratechnologies, and ViiV Healthcare; and payment for speaker's bureaus from Gilead Sciences, Inc.

Acknowledgments: We thank all study participants, study investigators, and staff. This study was funded by Gilead Sciences, Inc. Medical writing support was provided by Anne Errichelli, DPhil, CMPP (Aspire Scientific Ltd, UK), and was funded by Gilead Sciences, Inc.

Correspondence: Karam Mounzer, mounzerk@fight.org.

Infectious Diseases Society of Ireland (IDSI) 2024; May 16-17, 2024; Dun Laoghaire, Ireland