Once-Weekly Islatravir Plus Lenacapavir in Virologically Suppressed People with HIV: Week 48 Safety Profile, Efficacy, and Metabolic Changes

Amy E. Colson[1], Gordon E. Crofoot[2], Peter J. Ruane[3], Moti N. Ramgopal[4], Alexandra W. Dretler[5], Ronald G. Nahass[6], Gary I. Sinclair[7], Mezgebe Berhe[8], Fadi Shihadeh[9], Shan-Yu Liu[9], Stephanie Klopfer[10], Sharline Madera[9], Hadas Dvory-Sobol[9], Martin S. Rhee[9], Elizabeth G. Rhee[10], Conor Moran[11]*, Jared Baeten[9], Joseph Eron[12]

[1]Community Resource Initiative, Boston, Massachusetts, USA; [2]The Crofoot Research Center, Houston, Texas, USA; [3]Ruane Clinical Research, Los Angeles, California, USA; [4]Midway Immunology & Research Center, Fort Pierce, Florida, USA; [5]Infectious Disease Specialists of Atlanta, Decatur, Georgia, USA; [6]IDCare, Hillsborough, New Jersey, USA; [7]Prism Health North Texas, Dallas, Texas, USA; [8]North Texas Infectious Diseases Consultants, Dallas, Texas, USA; [9]Gilead Sciences, Foster City, California, USA; [10]Merck & Co., Inc., Rahway, New Jersey, USA; [11]Gilead Sciences Ireland UC, Dublin, Ireland; [12]University of North Carolina, Chapel Hill, North Carolina, USA. *Listed as author for presentation purposes only with permission of all authors

Conclusions

- Weekly oral ISL+LEN maintained high rates of virologic suppression (94.2%) at Week 48 in people with HIV who were virologically suppressed
 - — No participant on ISL+LEN had HIV-1 RNA ≥50 c/mL at Week 48 or at study discontinuation
- Weekly oral ISL+LEN was well tolerated, as evidenced by the absence of any treatment-related Grade ≥3 AEs or serious AEs
- There were no between-group differences in CD4+ T-cell or lymphocyte count changes from baseline through Week 48
- There were no between-group differences in body weight or BMI changes from baseline through Week 48
- Participants demonstrated high rates (99.2%) of adherence to oral weekly ISL+LEN
- The Phase 2 results support advancing the weekly oral ISL+LEN regimen to Phase 3 trials: ISLEND-1 and ISLEND-2 (NCT06630286; NCT06630299)
- ISL + LEN has the potential to become the first oral weekly complete regimen for the treatment of HIV-1 infection

AE, adverse event; BMI, body mass index; c/mL, copies/ml; ISL, islatravir; LEN, lenacapavir;

Background

- Once-weekly (QW) oral antiretrovirals (ARVs) have the potential to address pill fatigue and adherence challenges related to daily oral treatment for HIV-1 infection[1]
- Islatravir (ISL) is a nucleoside reverse transcriptase translocation inhibitor[2]
 - — Prior ISL studies have shown dose/exposure-related decreases in CD4+ T-cell and lymphocyte counts[3]
 - Pharmacokinetic modelling indicates such declines are not expected with the 2 mg dose chosen for this study[4]
- Lenacapavir (LEN) is a first-in-class capsid inhibitor[5]
- Both ISL and LEN have multiple mechanisms of action, potent ARV activity at low doses, and long half-lives ($t_{1/2}$) that allow for QW dosing[6–8,a]
- Primary endpoint data (Week 24) from the current, ongoing Phase 2 study (NCT05052996) were previously reported
 - — Most participants (94.2%) maintained viral suppression in the QW oral ISL+LEN group[9]

[a]LEN $t_{1/2}$=10–12 days; ISL-triphosphate $t_{1/2}$=7–9 days.

Objective

- To investigate the efficacy and safety profile of QW oral ISL+LEN in people with HIV-1 who are virologically suppressed at Week 48

Methods

Study Design

A Phase 2, Open-label, Active-Controlled Study in People with HIV who are Virologically Suppressed

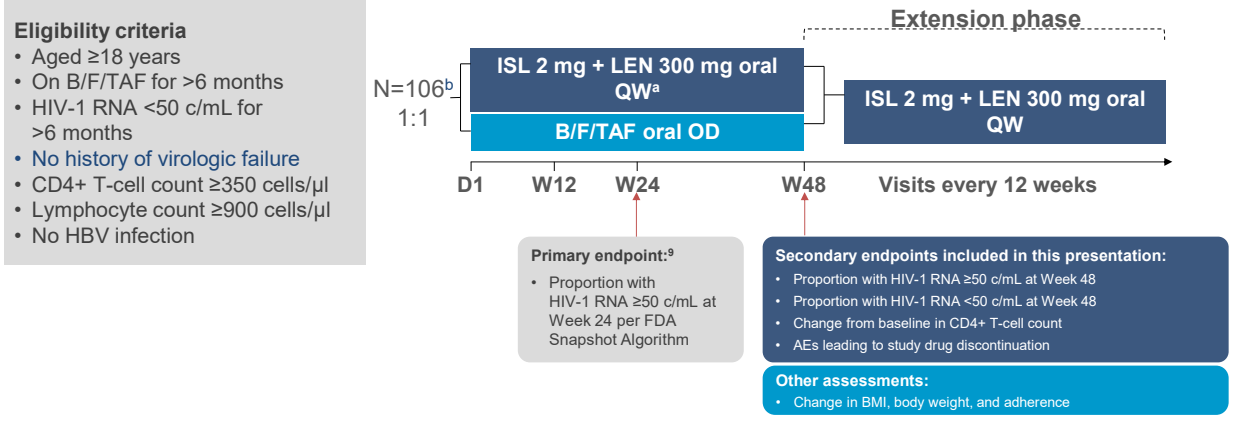

Eligibility criteria
• Aged ≥18 years
• On B/F/TAF for >6 months
• HIV-1 RNA <50 c/mL for >6 months
• No history of virologic failure
• CD4+ T-cell count ≥350 cells/µl
• Lymphocyte count ≥900 cells/µl
• No HBV infection
N=106[b]
1:1
ISL 2 mg + LEN 300 mg oral QW[a]
B/F/TAF oral OD
Extension phase
ISL 2 mg + LEN 300 mg oral QW
D1
W12
W24
W48
Visits every 12 weeks
Primary endpoint:[9]
• Proportion with HIV-1 RNA ≥50 c/mL at Week 24 per FDA Snapshot Algorithm
Secondary endpoints included in this presentation:
• Proportion with HIV-1 RNA ≥50 c/mL at Week 48
• Proportion with HIV-1 RNA <50 c/mL at Week 48
• Change from baseline in CD4+ T-cell count
• AEs leading to study drug discontinuation
Other assessments:
• Change in BMI, body weight, and adherence

[a]600 mg of LEN was given on Day 1 and Day 2 for pharmacologic loading. [b]Randomised, N=106; dosed, n=104.
AE, adverse event; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BMI, body mass index; c/mL, copies/ml; D, Day; FDA, Food and Drug Administration; HBV, hepatitis B virus; ISL, islatravir; LEN, lenacapavir; OD, daily; QW, weekly; W, Week.

Results

Baseline Demographic and Disease Characteristics

	ISL+LEN (n=52)	B/F/TAF (n=52)	Total (N=104)
Median (range) age, years	40 (28–67)	40 (26–76)	40 (26–76)
Assigned female at birth, n (%)	10 (19.2)	9 (17.3)	19 (18.3)
Gender identity, n (%)			
Transgender female	1 (1.9)	0	1 (1.0)
Non-binary/third gender	0	1 (1.9)	1 (1.0)
Race, n (%)			
White	25 (48.1)	27 (51.9)	52 (50.0)
Black	21 (40.4)	16 (30.8)	37 (35.6)
Asian	2 (3.8)	1 (1.9)	3 (2.9)
American Indian or Alaska Native	1 (1.9)	2 (3.8)	3 (2.9)
Native Hawaiian or Pacific Islander	0 (0)	1 (1.9)	1 (1.0)
Other	3 (5.8)	5 (9.6)	8 (7.7)
Hispanic or Latinx ethnicity, n (%)	13 (25.0)	17 (32.7)	30 (28.8)
Mean (SD) CD4+ T-cell count, cells/µL	755 (223.6)	818 (271.3)	786 (249.5)
Mean (SD) lymphocyte count x 10^3 cells/µL	1.94 (0.445)	1.95 (0.652)	1.94 (0.556)
Median (IQR) body weight, kg	79.3 (70.4–87.4)	83.2 (76.1–92.5)	80.5 (74.4–88.7)
Median (IQR) BMI, kg/m²	26.9 (23.8–30.0)	27.2 (25.5–29.3)	27.1 (24.5–29.4)

B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BMI, body mass index; IQR, interquartile range; ISL, islatravir; LEN, lenacapavir.

Results

Virologic Outcomes at Week 48 by FDA Snapshot Algorithm

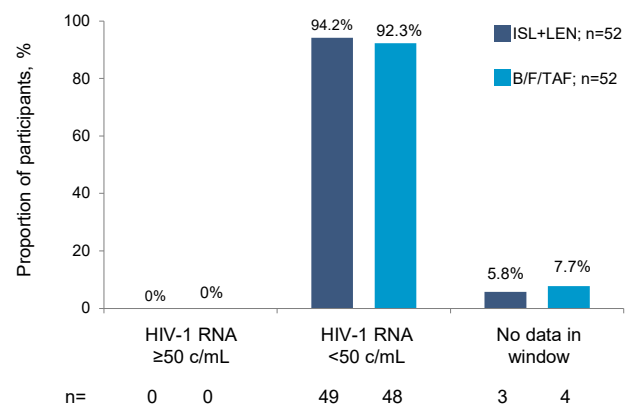

Proportion of participants, %
100
80
60
40
20
0
ISL+LEN; n=52
B/F/TAF; n=52
0%
0%
94.2%
92.3%
5.8%
7.7%
HIV-1 RNA ≥50 c/mL
HIV-1 RNA <50 c/mL
No data in window
n=
0
0
49
48
3
4

Participants with no data in window:

ISL+LEN (n=3)
- Two participants discontinued due to AEs not related to study drug
- One participant discontinued due to other reasons not related to study drug
- All participants had HIV-1 RNA <50 c/mL at study discontinuation

B/F/TAF (n=4)
- Three participants discontinued due to other reasons not related to study drug and had HIV-1 RNA <50 c/mL at study discontinuation
- One participant had missing data during window, but remained on study drug

Participants in both treatment groups maintained high rates of virologic suppression

AE, adverse event; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c/mL, copies/mL; FDA, Food and Drug Administration; ISL, islatravir; LEN, lenacapavir.

Adverse Events

Participants, n (%)	ISL+LEN (n=52)	B/F/TAF (n=52)
Any AE	42 (80.8)	40 (76.9)
Treatment-related AE	10 (19.2)	3 (5.8)
Grade 1 or 2	10 (19.2)	3 (5.8)
≥2 participants in ISL+LEN group		
Dry mouth	2 (3.8)	0
Nausea	2 (3.8)	0
Grade 3 or 4	0	0
Serious AE	3 (5.8)[a]	0
Treatment-related	0	0
AE leading to study drug discontinuation	2 (3.8)[b]	0
Treatment-related	0	0

No Grade 3 or higher AEs, serious AEs, or AEs leading to discontinuation were considered related to the study drug by the investigator

[a]Serious AEs included large intestine perforation and renal colic (in the same participant), pneumonia, and neurologic anesthetic complication. [b]Large intestine perforation and renal colic, n=1; acute hepatitis B infection, n=1 (both participants had HIV-1 RNA <50 c/mL at study discontinuation).
AE, adverse event; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c/mL, copies/mL; ISL, islatravir; LEN, lenacapavir.

Laboratory Anomalies

Laboratory abnormalities occurring in ≥1 participant in the ISL+LEN group, n/N (%)	ISL+LEN (n=52)	B/F/TAF (n=52)
Grade 3		
Creatinine (increased)	1/52 (1.9)	0/51
Creatinine clearance (decreased)	2/52 (3.8)	2/51 (3.9)
Non-fasting hyperglycaemia	1/43 (2.3)	2/43 (4.7)
Glycosuria[a]	1/52 (1.9)	2/51 (3.9)
Hyperkalaemia	1/52 (1.9)	0/51
ALT (increased)[b]	1/52 (1.9)	0/51
Grade 4		
Creatine kinase (increased)[c]	2/52 (3.8)	0/51

No Grade 3 and 4 laboratory abnormalities were clinically significant, except ALT elevation seen in a participant with acute hepatitis B

[a]Glycosuria occurred in participants with type 2 diabetes mellitus. [b]Increased ALT occurred in the participant with acute hepatitis B. [c]Increased creatinine kinase occurred after vigorous exercise in both participants.
ALT, alanine transaminase; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; ISL, islatravir; LEN, lenacapavir.

CD4+ T-cell and Lymphocyte Count Changes Through Week 48

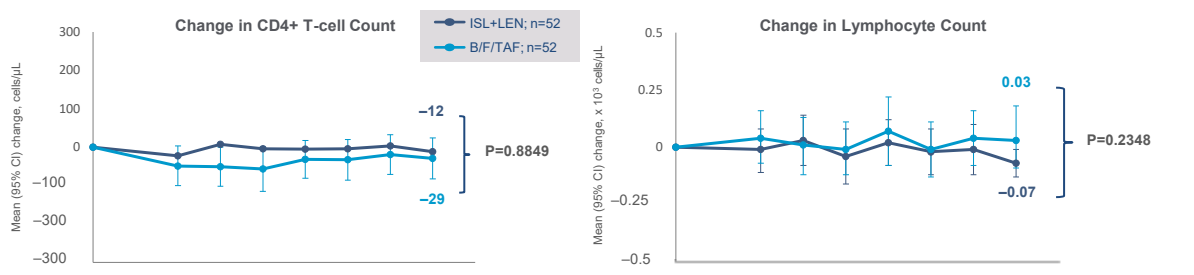

Change in CD4+ T-cell Count
ISL+LEN; n=52
B/F/TAF; n=52
Mean (95% CI) change, cells/µL
300
200
100
0
−100
−300
−300
−12
−29
P=0.8849
Change in Lymphocyte Count
Mean (95% CI) change, x 10³ cells/µL
0.5
0.25
0
−0.25
−0.5
0.03
−0.07
P=0.2348

Mean Values	BL	W12	W18	W24	W30	W36	W42	W48
ISL+LEN	755	732	766	755	754	756	761	746
B/F/TAF	818	758	767	761	785	783	797	787

Mean Values	BL	W12	W18	W24	W30	W36	W42	W48
ISL+LEN	1.94	1.94	1.98	1.92	1.96	1.94	1.93	1.88
B/F/TAF	1.95	1.99	1.97	1.96	2.03	1.96	2.00	1.99

- **There were no significant differences between groups in mean change from baseline in CD4+ T-cell or lymphocyte counts at Week 48**
- **No participants discontinued due to a decrease in CD4+ T-cell or lymphocyte counts**

B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BL, baseline; ISL, islatravir; LEN, lenacapavir; W, Week.

Body Weight and BMI Changes Through Week 48

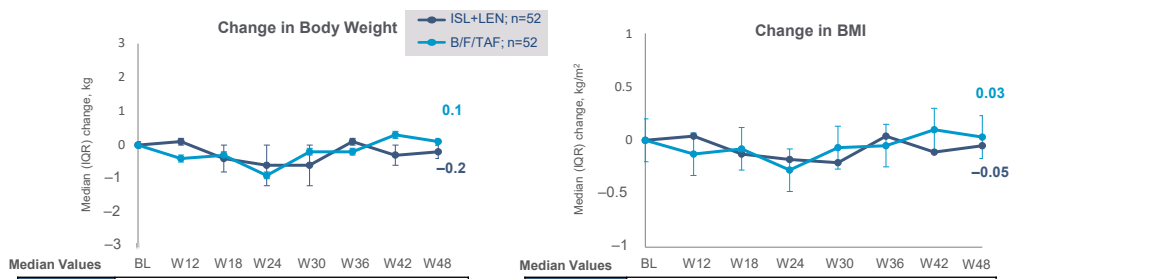

Change in Body Weight
ISL+LEN; n=52
B/F/TAF; n=52
Median (IQR) change, kg
3
2
1
0
−1
−2
−3
0.1
−0.2
Change in BMI
Median (IQR) change, kg/m²
1
0.5
0
−0.5
−1
0.03
−0.05

Median Values	BL	W12	W18	W24	W30	W36	W42	W48
ISL+LEN	79.3	79.1	77.2	77.9	77.7	78.8	77.9	77.6
B/F/TAF	83.2	83.4	82.8	82.0	84.7	84.7	84.2	84.0

Median Values	BL	W12	W18	W24	W30	W36	W42	W48
ISL+LEN	26.9	26.8	26.9	26.6	27.2	27.1	26.6	26.7
B/F/TAF	27.2	27.4	27.3	27.3	27.4	27.9	27.6	27.7

No between-group differences in median change in body weight and BMI at Week 48

B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BL, baseline; BMI, body mass index; IQR, interquartile range; ISL, islatravir; LEN, lenacapavir; W, Week.

Adherence (by Pill Count) Through Week 48

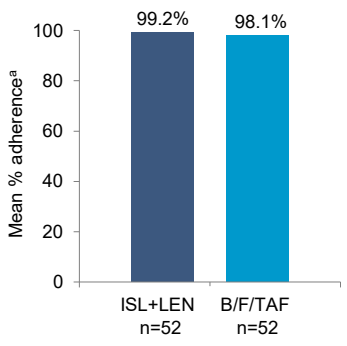

Mean % adherence[a]
100
80
60
40
20
0
99.2%
98.1%
ISL+LEN n=52
B/F/TAF n=52

Adherence was high for ISL+LEN and B/F/TAF through Week 48

[a]Adherence (%) calculated as: (total pills taken / total pills prescribed) x 100. Denominator for the adherence rate category summaries was based on number of participants who returned at least 1 bottle and had calculable adherence. For ISL, 2 capsules taken (1 mg each) was considered as a pill.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; ISL, islatravir; LEN, lenacapavir.

References: **1.** Claborn KR, et al. *Psychol Health Med* 2015;20:255–65. **2.** Schürmann D, et al. *Lancet HIV* 2020;7:e164–72. **3.** Squires K, et al. CROI 2023; Abstract 192. **4.** Vargo RC, et al. CROI 2023; Poster 497. **5.** Sunlenca® Prescribing Information, available at https://www.gilead.com/-/media/files/pdfs/medicines/hiv/sunlenca/sunlenca_pi.pdf (accessed November 2024). **6.** Zhang H, et al. CROI 2022; Abstract 433. **7.** Shaik N, et al. AIDS 2022; Poster PESUB23. **8.** Matthews R, et al. *Clin Trans Sci* 2021;14:1935–44. **9.** Colson A, et al. CROI 2024; Abstract 208.

Acknowledgments: We extend our thanks to the participants and their families. We extend our thanks to all the participating investigators: Shauna Applin, Archana Asundi, Paul Benson, Mezgebe Berhe, Cynthia Brinson, Larry M. Bush, Amy E. Colson, Catherine M. Creticos, Gordon E. Crofoot, Edwin DeJesus, Alexandra W. Dretler, Joseph Eron, Cynthia Firnhaber, Edward Gardner, Linda Gorgos, Debbie Hagins, Shawn Hassler, Theo Hodge, Dushyantha Jayaweera, Ronald G. Nahass, Moti N. Ramgopal, Gary J. Richmond, Afsoon Roberts, Peter J. Ruane, Rachel Safran, Laura Salazar, William Sanchez, Patric Schine, Sorana Segal-Maurer, Peter Shalit, Cecilia M. Shikuma, Gary I. Sinclair, Christine Zurawski, Marcus Tellez, Kimberly Workowski. This study was funded by Gilead Sciences Inc, Foster City, CA, USA and is part of a collaboration between Gilead Sciences Inc., Foster City, CA, USA and Merck Sharp & Dohme LLC., a subsidiary of Merck & Co., Inc., Rahway, NJ, USA. All authors contributed to and approved the presentation. Medical writing support was provided by Bill Wang of Gilead Sciences, Inc. Editorial support was provided by Ashfield MedComms, an Inzio company, and was funded by Gilead Sciences, Inc.

Correspondence: Dr Amy Colson, acolson@crihealth.org

Disclosures: **AC,** Gilead Sciences, Inc. (consulting fees); ViiV (honoraria). **GC,** Gilead Sciences, Inc. (grant/research support); ViiV (grant/research support); Janssen (grant/research support); Merck (grant/research support); AbbVie (grant/research support). **PR,** Gilead Sciences, Inc. (advisor/consultant, honoraria); ViiV (advisor/consultant, honoraria). **MR,** Gilead Sciences, Inc. (advisor/consultant, honoraria); ViiV (advisor/consultant, honoraria); Merck (advisor/consultant); AbbVie (honoraria). **AD,** Gilead Sciences, Inc. (grant/research support); ViiV (grant/research support, advisor/consultant); AbbVie (grant/research support). **RN,** Merck, Vir, Arbutus, Gilead Sciences, Inc., Insmed (grants). **GS,** Gilead Sciences, Inc. (advisor/consultant; grant/research support); Janssen (advisor/consultant; grant/research support; honoraria); ViiV (advisor/consultant, grant/research support, honoraria); Theratechnologies (advisor/consultant, grant/research support, honoraria); Merck (advisor/consultant, grant/research support, honoraria); Abbvie (grant/research support). **MB,** none. **FS, SL, SM, HD-S, MR,** and **JB** are all employees and shareholders of Gilead Sciences, Inc. **SK** and **ER** are employees of Merck Sharp & Dohme LLC., a subsidiary of Merck & Co., Inc., Rahway, New Jersey, USA; and shareholders of Merck & Co., Inc., Rahway, New Jersey, USA. **JE,** reports grants/contract payments made to his institution from ViiV and Gilead Sciences, Inc.; consulting fees from Gilead Sciences, Inc., ViiV, Merck, and Abbvie; and fees from Invivyd and Taimed (DSMB/advisory board). **SK,** full time employee of Gilead Sciences Ireland UC and shareholder of Gilead Sciences, Inc.

IDSI 2025, 15-16 May 2025, Dun Laoghaire, Ireland